Effects of inhaled PC945 on fungal load in mouth wash collected from healthy subjects in the first-in-human study (NCT02715570)

L. Daly, K. Woodward, M. Coates, L. Cass, P. Strong, A. Murray, G. Rapeport and K. Ito

Pulmocide Ltd, London, United Kingdom

Introduction/Aim

A first in human (FIH) study of PC945, a novel inhaled antifungal triazole[1-3], was conducted to evaluate the safety and pharmacokinetics of PC945 delivered as a suspension formulation by nebulisation (NCT02715570, EudraCT Number: 2015-003327-64). During this study, mouth wash and pharyngeal swabs were collected for exploratory investigation of the impact of PC945 treatment on oropharyngeal fungal loads.

Methods

Healthy volunteers (HVs) received PC945 as either single inhaled ascending doses (0.5-10 mg) or placebo (SAD Cohort) or 5 mg once daily for 7 days ([RD] cohort). Patients with mild asthma received a single inhaled dose of PC945, 5 mg or Placebo (asthma cohort). PC945 was well tolerated following doses in healthy subjects and patients with mild asthma. Mouth wash and pharyngeal swabs were collected pre- and 48hrs post dose. Mouth wash samples were cultured on PGC agar (Potato Dextrose Agar with chloramphenicol and gentamycin) and CHROMAgar, and the presence of *Candida albicans/Aspergillus fumigatus* assessed using polymerase chain reaction (PCR). Yeasts/Moulds were identified using CHROMAgar, PCR or Maldi-Tof MS (BCCM/IHEM) analyses.

Results

Yeast

Cohort 1: Single ascending dose (SAD)
(2 placebo + 6 at each of 4 PC945 doses)

72% of pre-dose samples (23/32) were yeast positive on PGC agar and 66% of samples (21/32) were yeast positive on CHROMagar. The most frequently observed yeast was *Candida albicans*. *Candida parapsirosis*, *Candida krusei* and *Candida inconpukia* were also observed occasionally.

PC945 dose-dependently inhibited yeast load detected on PGC agar and CHROMagar up to the 5mg dose. However, one subject (PC945 10mg group) had *Candida parapsilosis* infection, which seemed to be unaffected by PC945. Overall, treatment with PC945 statistically significantly inhibited *Candida* culture load (p=0.023) and PCR signal (p=0.009) (paired comparison pre- versus post-dose) in samples yeast positive pre-dose (Figure 1).

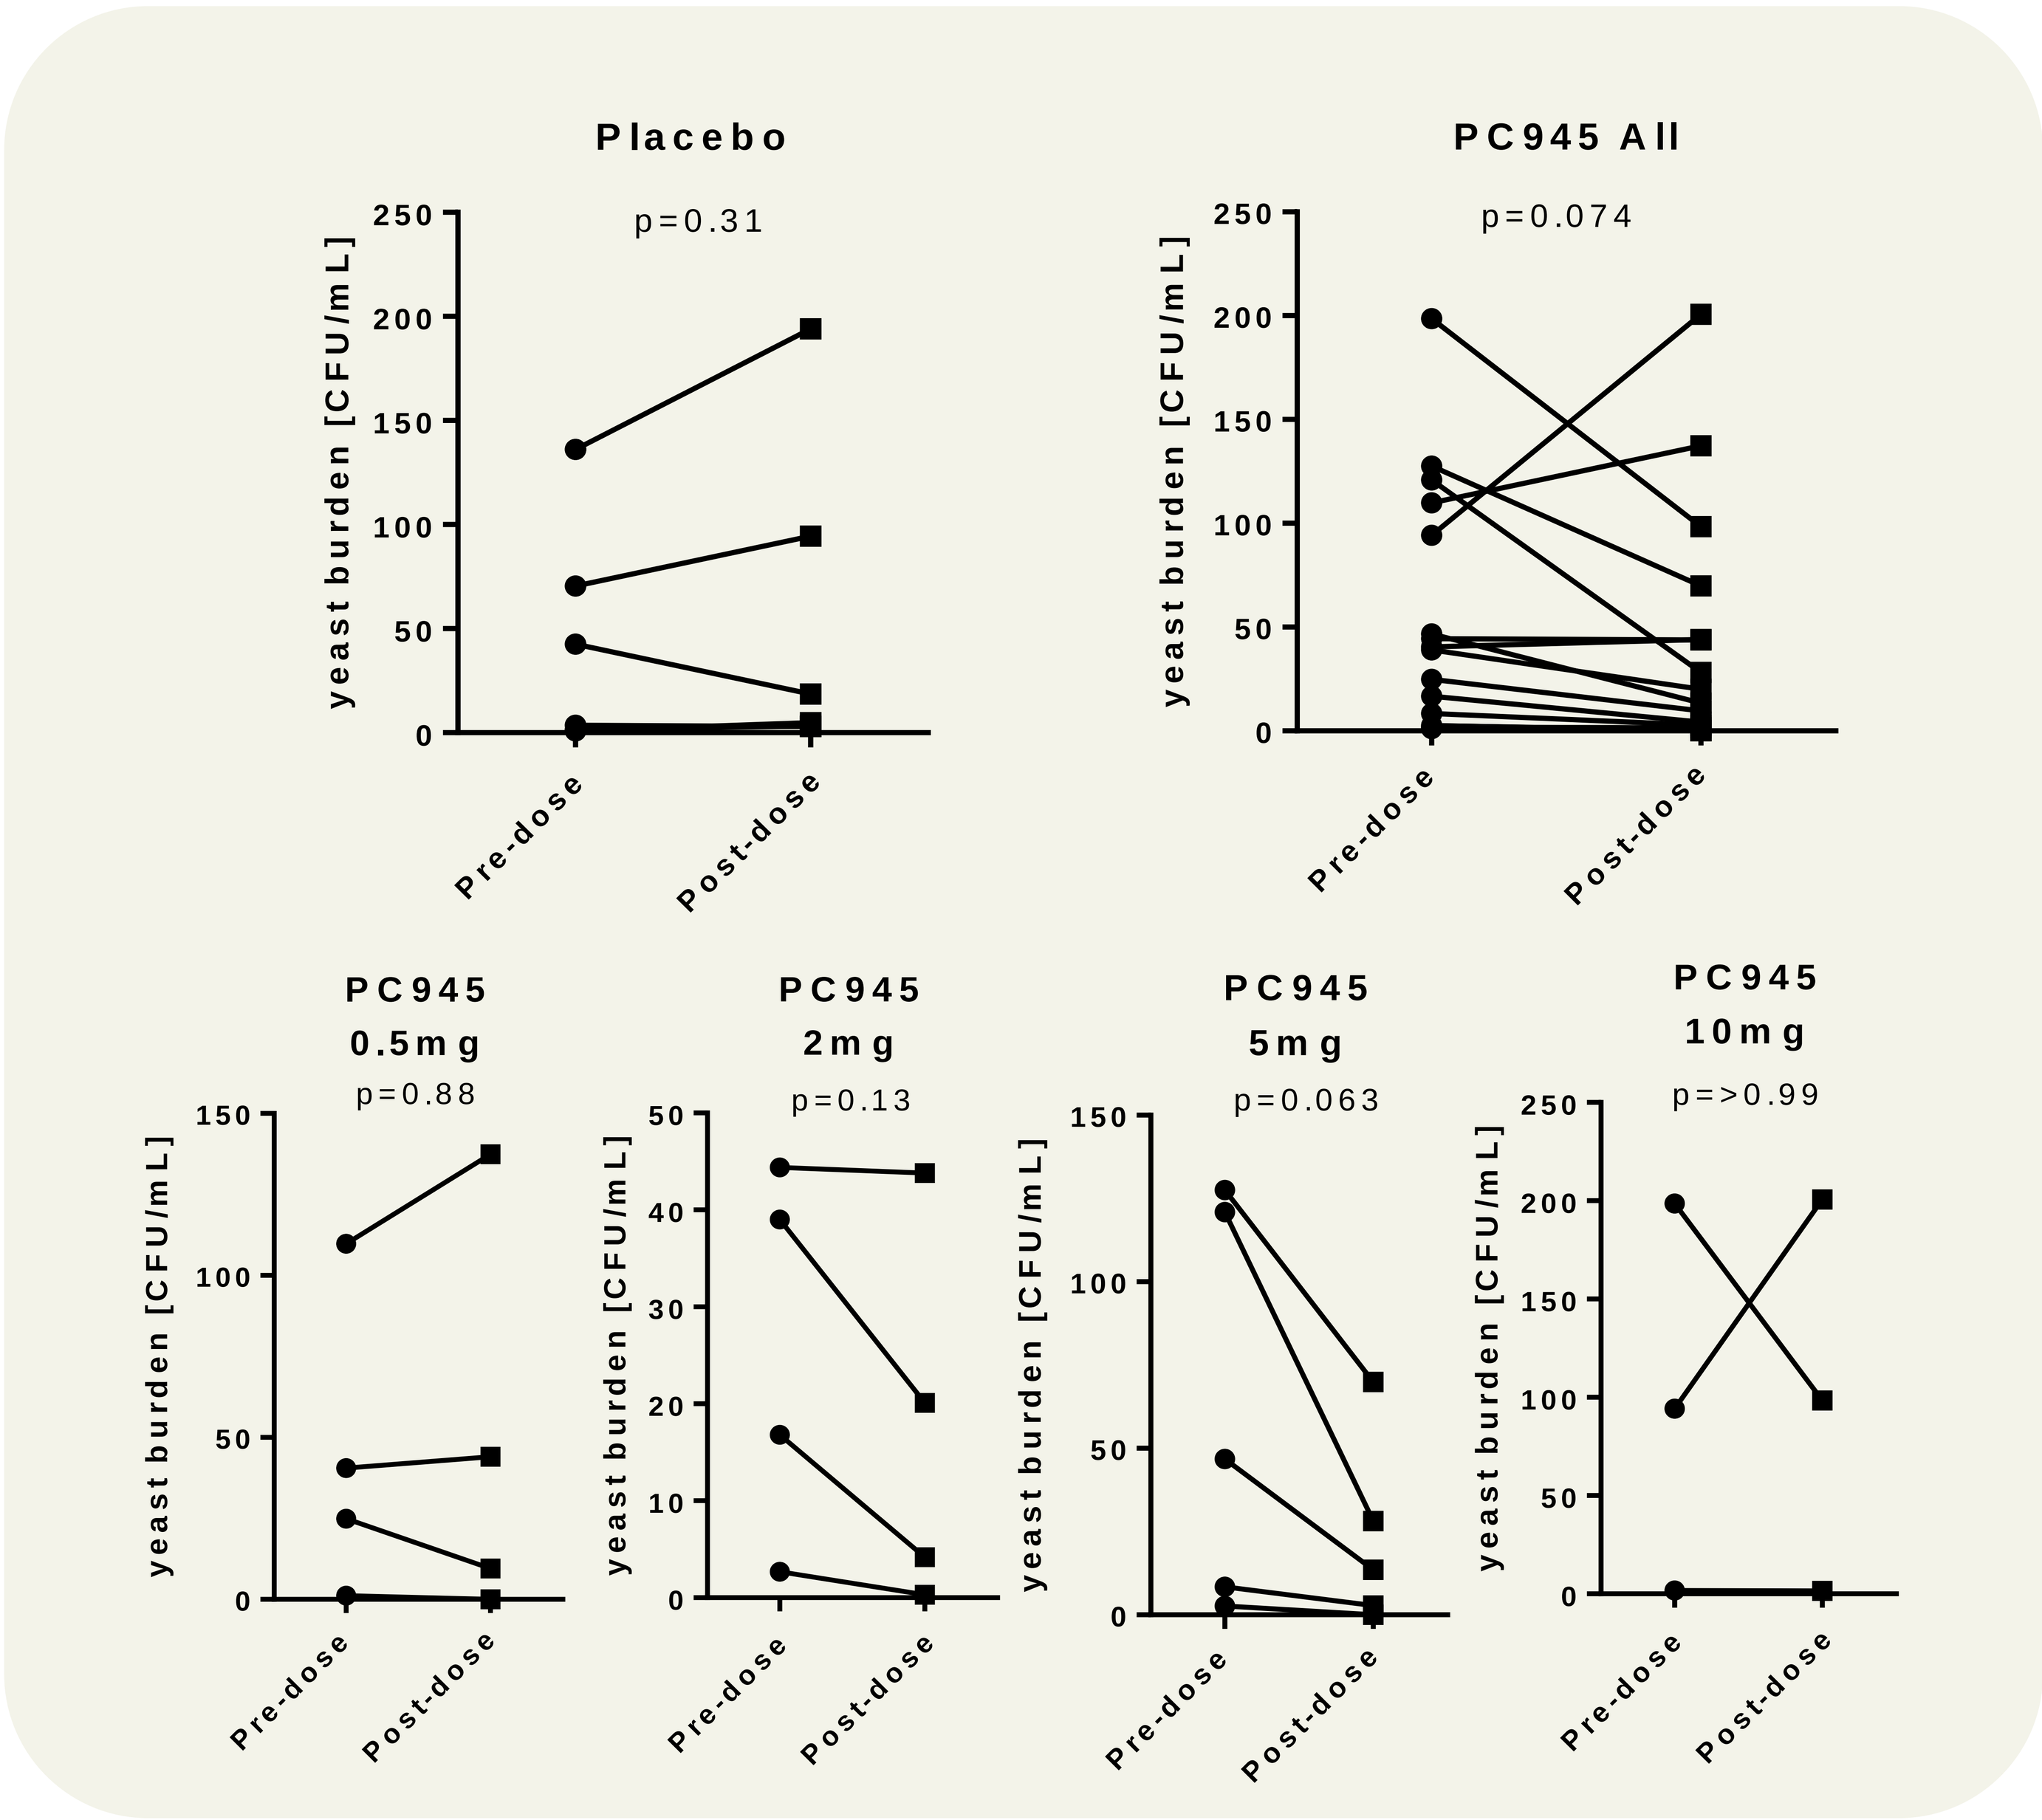
Placebo
p=0.31
PC945 All
p=0.074
PC945 0.5mg
p=0.88
PC945 2mg
p=0.13
PC945 5mg
p=0.063
PC945 10mg
p=>0.99
yeast burden [CFU/mL]
Pre-dose
Post-dose

Figure 1. Individual Yeast burden on PGC agar in Cohort 1 (samples with +ve culture at pre-dose only)

The PCR signals of *C. albicans* were slightly increased in Placebo samples after treatment (CT value was reduced), but PC945 treatment (>2mg) consistently inhibited the *C. albicans* PCR signal and, overall, PC945 statistically significantly inhibited the *C. albicans* PCR signal (p=0.019) where Placebo did not. For any samples with a CT=>35, the value of 35 was applied for statistical analysis purposes (Figure 2).

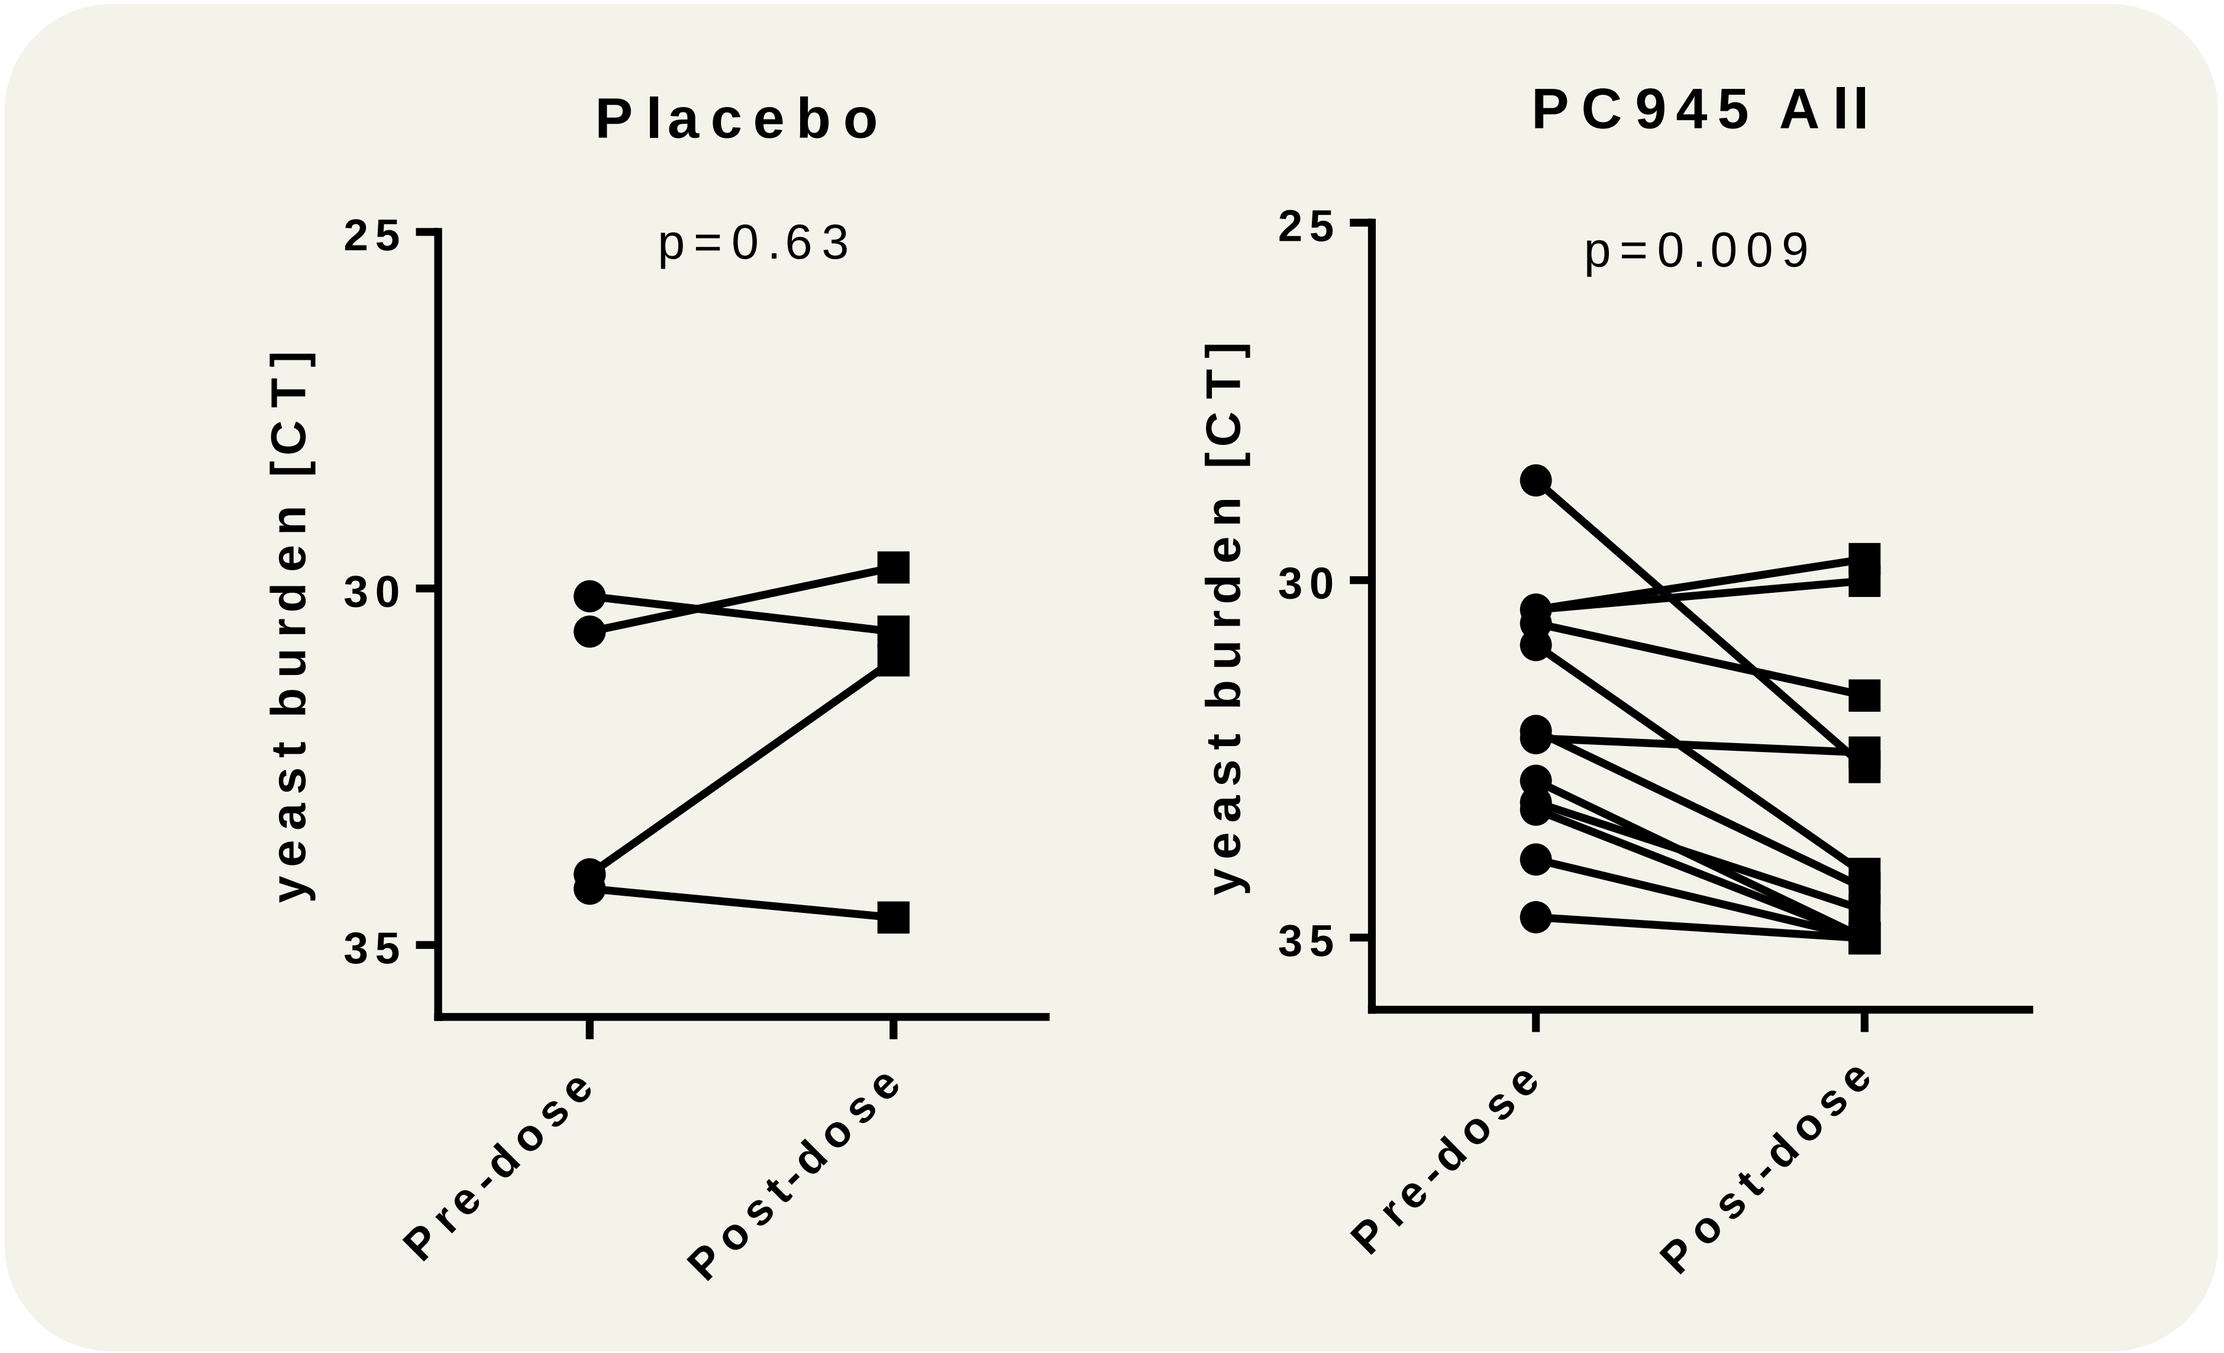
Placebo
p=0.63
PC945 All
p=0.009
yeast burden [CT]
Pre-dose
Post-dose

Figure 2. Individual plots of *C. albicans* PCR signal (for samples PCR +ve (CT: <35) at pre-dose only)

Cohort 2: Repeated dose (RD) (3 placebo + 6 PC945)

No sample in the placebo group was yeast culture positive (0/3) and only 2 samples out of 6 subjects in the PC945 treatment group were yeast culture positive pre-dose. Thus, fungal burden was low, so no meaningful analysis could be conducted.

Cohort 3: Single dose in patients with mild asthma (3 placebo + 6 PC945)

No yeast was detected in the Placebo treated group, but the trend of PC945 inhibition of yeasts at post-dose was observed compared with yeasts at pre-dose (Figure 3).

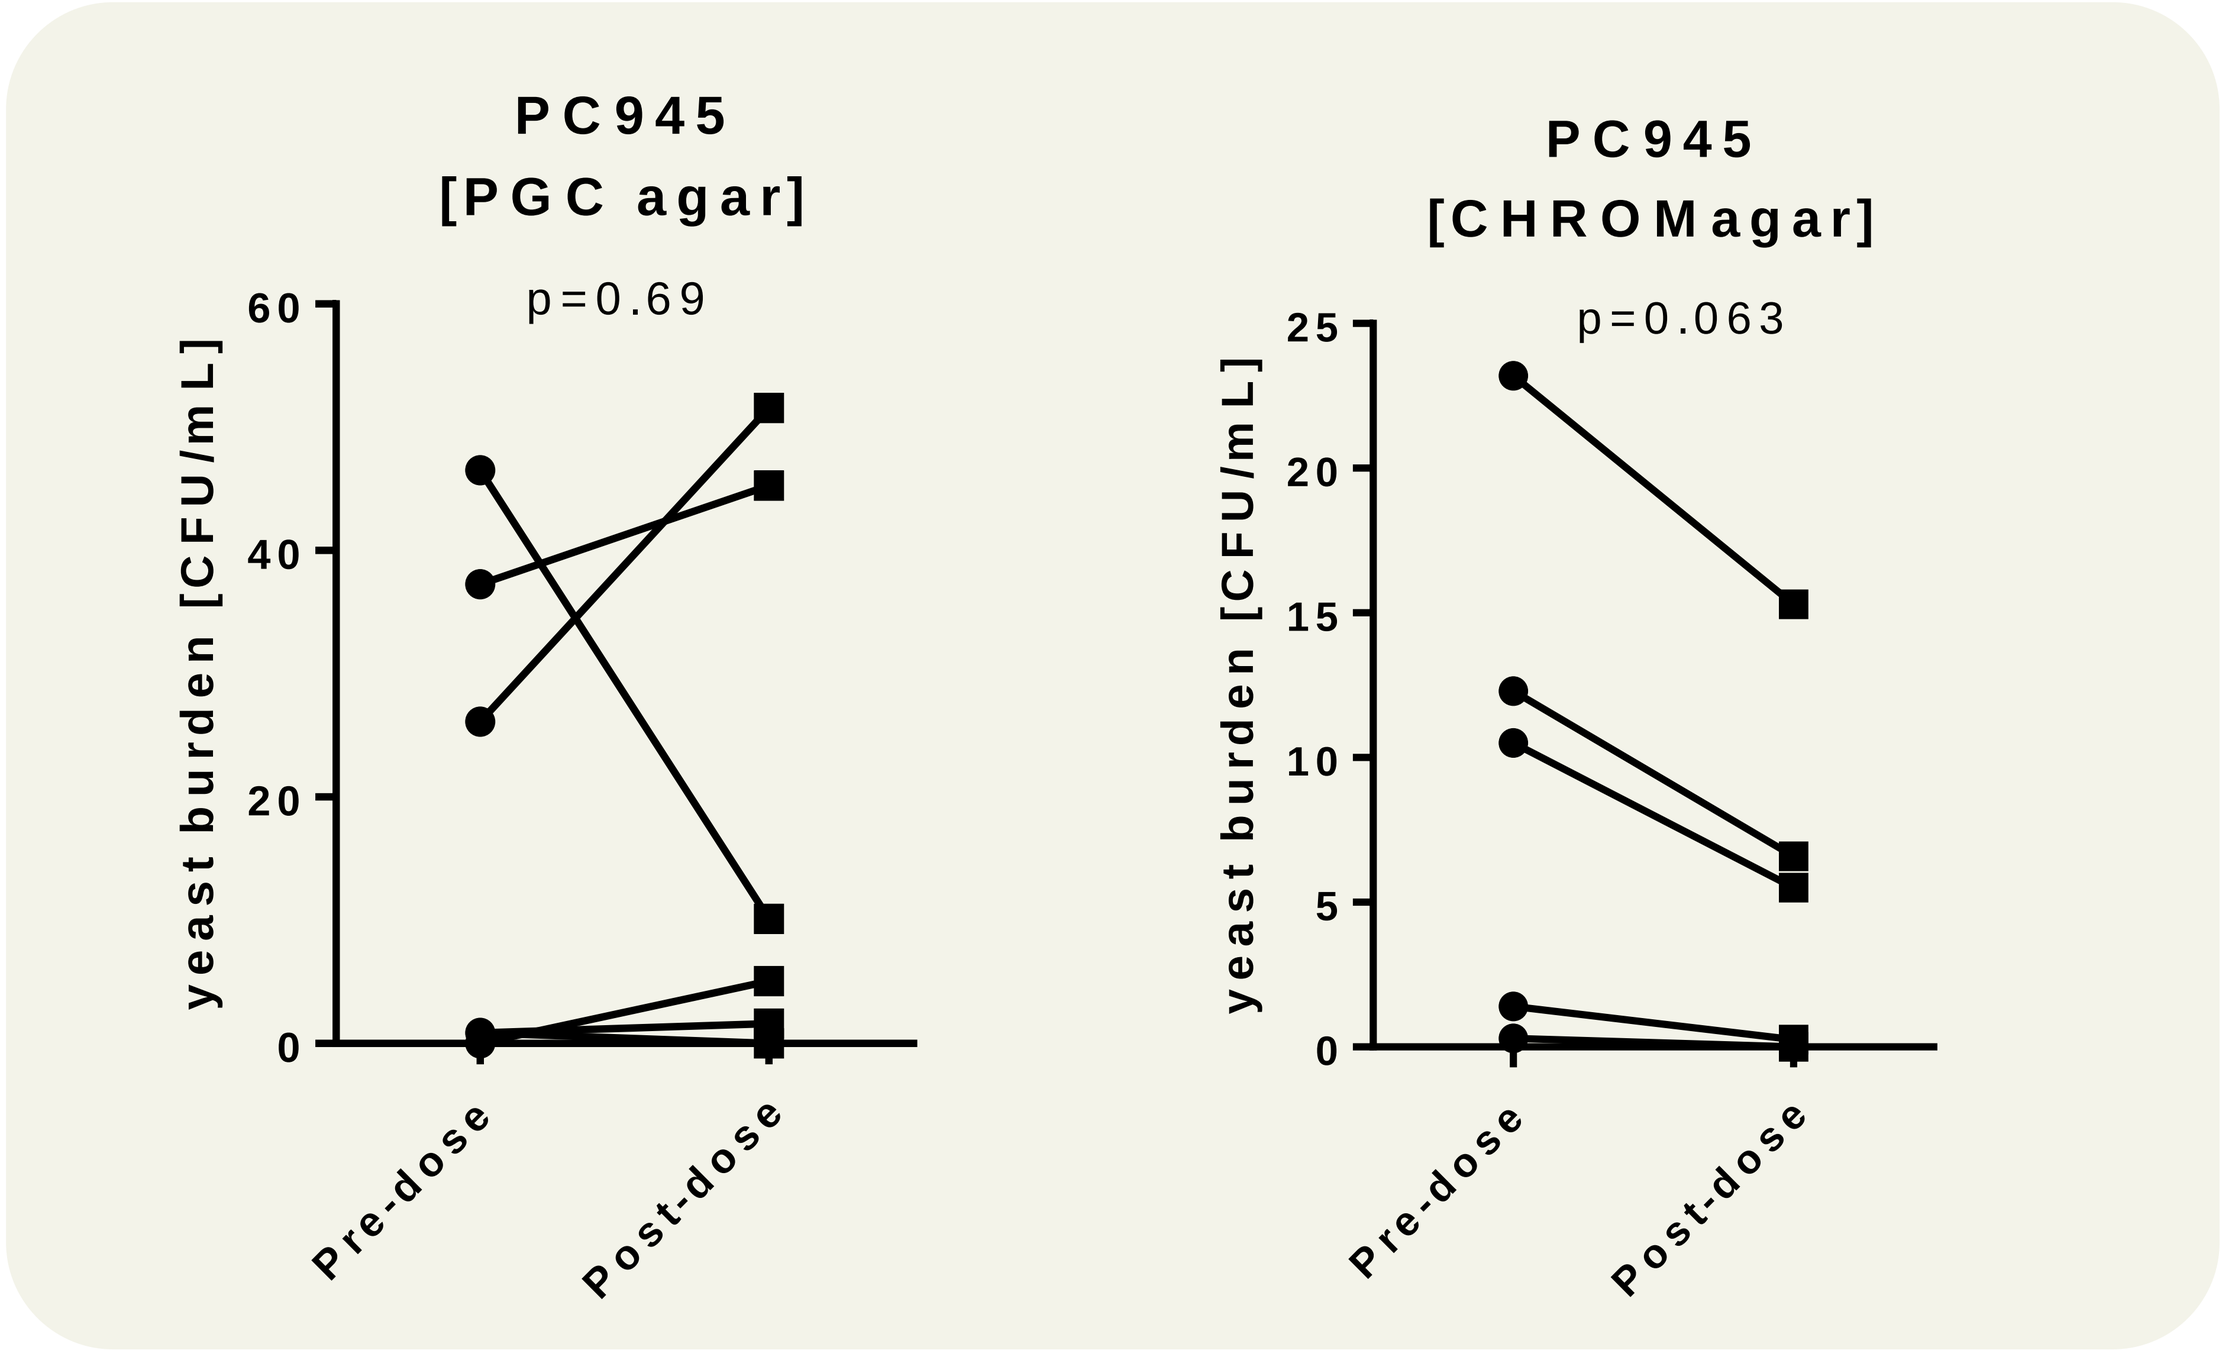
PC945
[PGC agar]
p=0.69
PC945
[CHROMagar]
p=0.063
yeast burden [CFU/mL]
Pre-dose
Post-dose

Figure 3 Individual plots of yeast burden in mouth wash on PGC agar (left) and CHROMagar (right) in Cohort 3 (subjects yeast culture positive samples pre-dose only)

Mould

In all cohorts, mould detection was limited (only 5 cases, low burden). The mould species observed were *Aspergillus fumigatus*, *Penicillium rubens* and *Penicillium chrysogenum*.

Despite limited mould detection in culture, 44% of samples (4/9) showed very weak PCR signals for *A. fumigatus* pre-dose in the asthma cohort. Based on CT values, a trend towards inhibition by PC945 was observed (Figure 5).

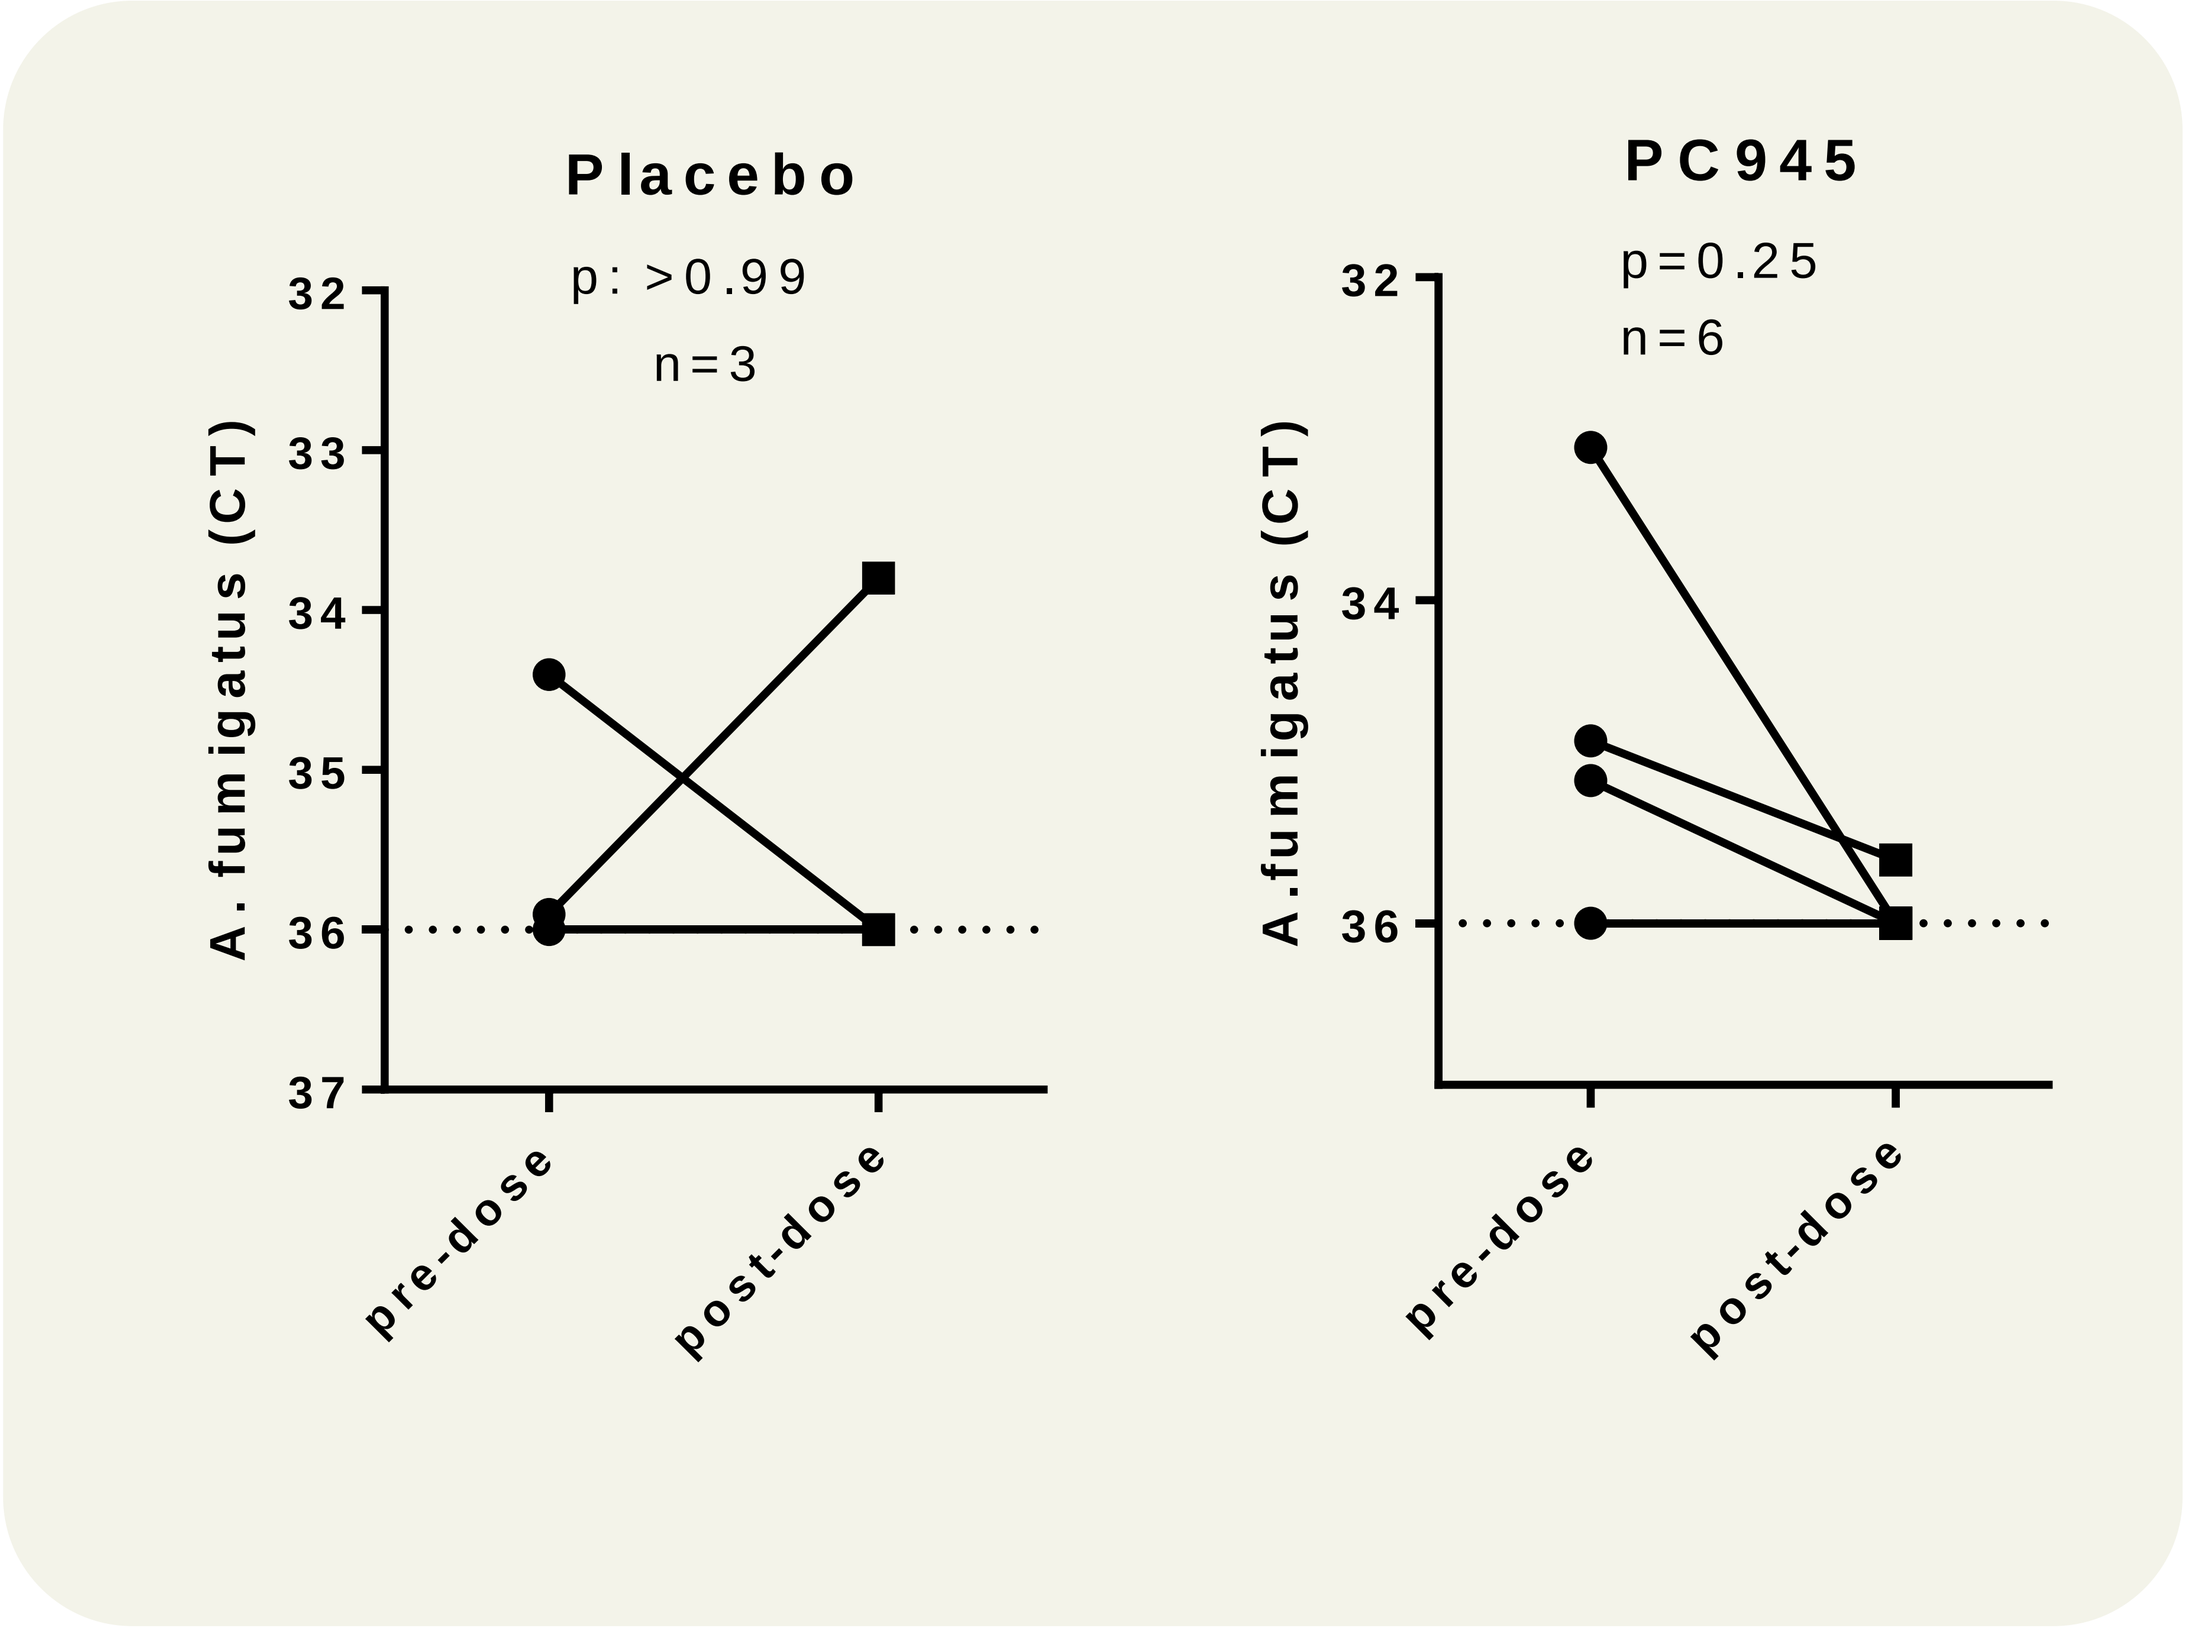
Placebo
p: >0.99
n=3
PC945
p=0.25
n=6
A. fumigatus (CT)
pre-dose
post-dose

Conclusion

Samples taken during this FIH study allowed a first exploratory investigation of PC945's antifungal activity in humans. Fungal detection rates and fungal burden in both cohorts of healthy volunteers were very low. In addition, fungal burden distribution was poorly randomised. Thus the study was neither powered nor appropriately randomised to assess potential treatment effects. The results do, however, suggest a trend of PC945 treatment-dependent inhibition of oral yeasts which may warrant further clinical study.

Reference

1. Colley T et al., Antimicrob Agents Chemother. 2017;61(5). pii: e02280-16.
2. Kimura G et al., Antimicrob Agents Chemother. 2017;61(9). pii: e00124-17.
3. Colley T et al., Sci Rep. 2019;9(1):9482.

PRINTED BY SCIENCEPOSTERS .co.uk